Bayesian Joint Modelling of Benefit and Risk in Drug Development

Maria Costa[1] & Thomas Drury[2]
[1] GSK, Statistical Innovation Group – Now working at Novartis
[2] Integral Statistics – Now working at GSK, Advanced Biostatistics and Data Analytics Centre of Excellence

PSI-2018

Introduction

- Understanding the benefit risk (BR) balance for treatments is critical in drug development.
- Sponsors and regulators agree that communication of BR assessments requires a structured approach, focusing on **key efficacy and safety outcomes**.
- A challenge in BR assessments is the potential for efficacy and safety outcomes to be connected at the subject level.
- Simulation studies and a real data example explored two Bayesian methods to assess BR allowing for dependence and using clinically meaningful thresholds to create joint posterior probability estimates.

Methods – Bayesian Joint Models

Generalized Linear Mixed Models

- Generalized Linear Mixed Models (GLMMs) extend linear mixed models for non-Gaussian distributed variables and can be used for **multiple responses with different distributions**.
- Using shared random effects across the different response models allow for dependence between the responses, the form of which depends on the link functions(s) and the distributions for both the responses and random effects.

Copula Marginal Regression

- Copula functions allow univariate distributions to be combined into multivariate ones whilst preserving the characteristics of each univariate component.
- Copula Marginal Regression (CMR) uses copula functions to incorporate separate (marginal) regression models into a single joint model allowing for dependence.

Bayesian Decision Making for Benefits and Risk

- The BR profile of a drug can be investigated using clinically meaningful thresholds, representing **minimum improvements in efficacy** and **maximum increases in risk** for a particular comparison (e.g. Active vs Control).
- These are independent and set by the project team and can be used as clinical Go/No-go boundaries.
- Bayesian joint models are used with clinically meaningful thresholds to estimate the joint posterior probabilities:

$$\mathbf{P}\ (\boldsymbol{H_E(\mu_E)} \ \geq\ \boldsymbol{\Delta_E} \text{ **and** } \boldsymbol{H_S(\mu_S)} \ \leq\ \boldsymbol{\Delta_S} |\ \textbf{Data, Prior}) \quad (1)$$

where:
$H_E(\cdot)$ and $H_S(\cdot)$ are comparisons based on the mean efficacy and safety responses.
Δ_E and Δ_S represent particular efficacy and safety thresholds of interest.

- The posterior probability (1) can be seen as the Probability of Technical Success (POTS).
- Contour plots showing pairs (Δ_E, Δ_S) vs posterior probability in (1) can support decision making, providing different stakeholders (sponsors, regulators, HTA bodies and patients) with the opportunity to assess if the data supports their own benefit-risk perspective.

Simulation Study 1: Single Efficacy and Safety

- Both models were used to assess a continuous efficacy response with a binary safety outcome for a scenario where the team wants to assess whether an asset should progress on to the next phase of development.
- Both methods successfully modelled the outcomes allowing for dependence with no bias.
- Intuitive plots were created for communicating the POTS for clinically meaningful thresholds.

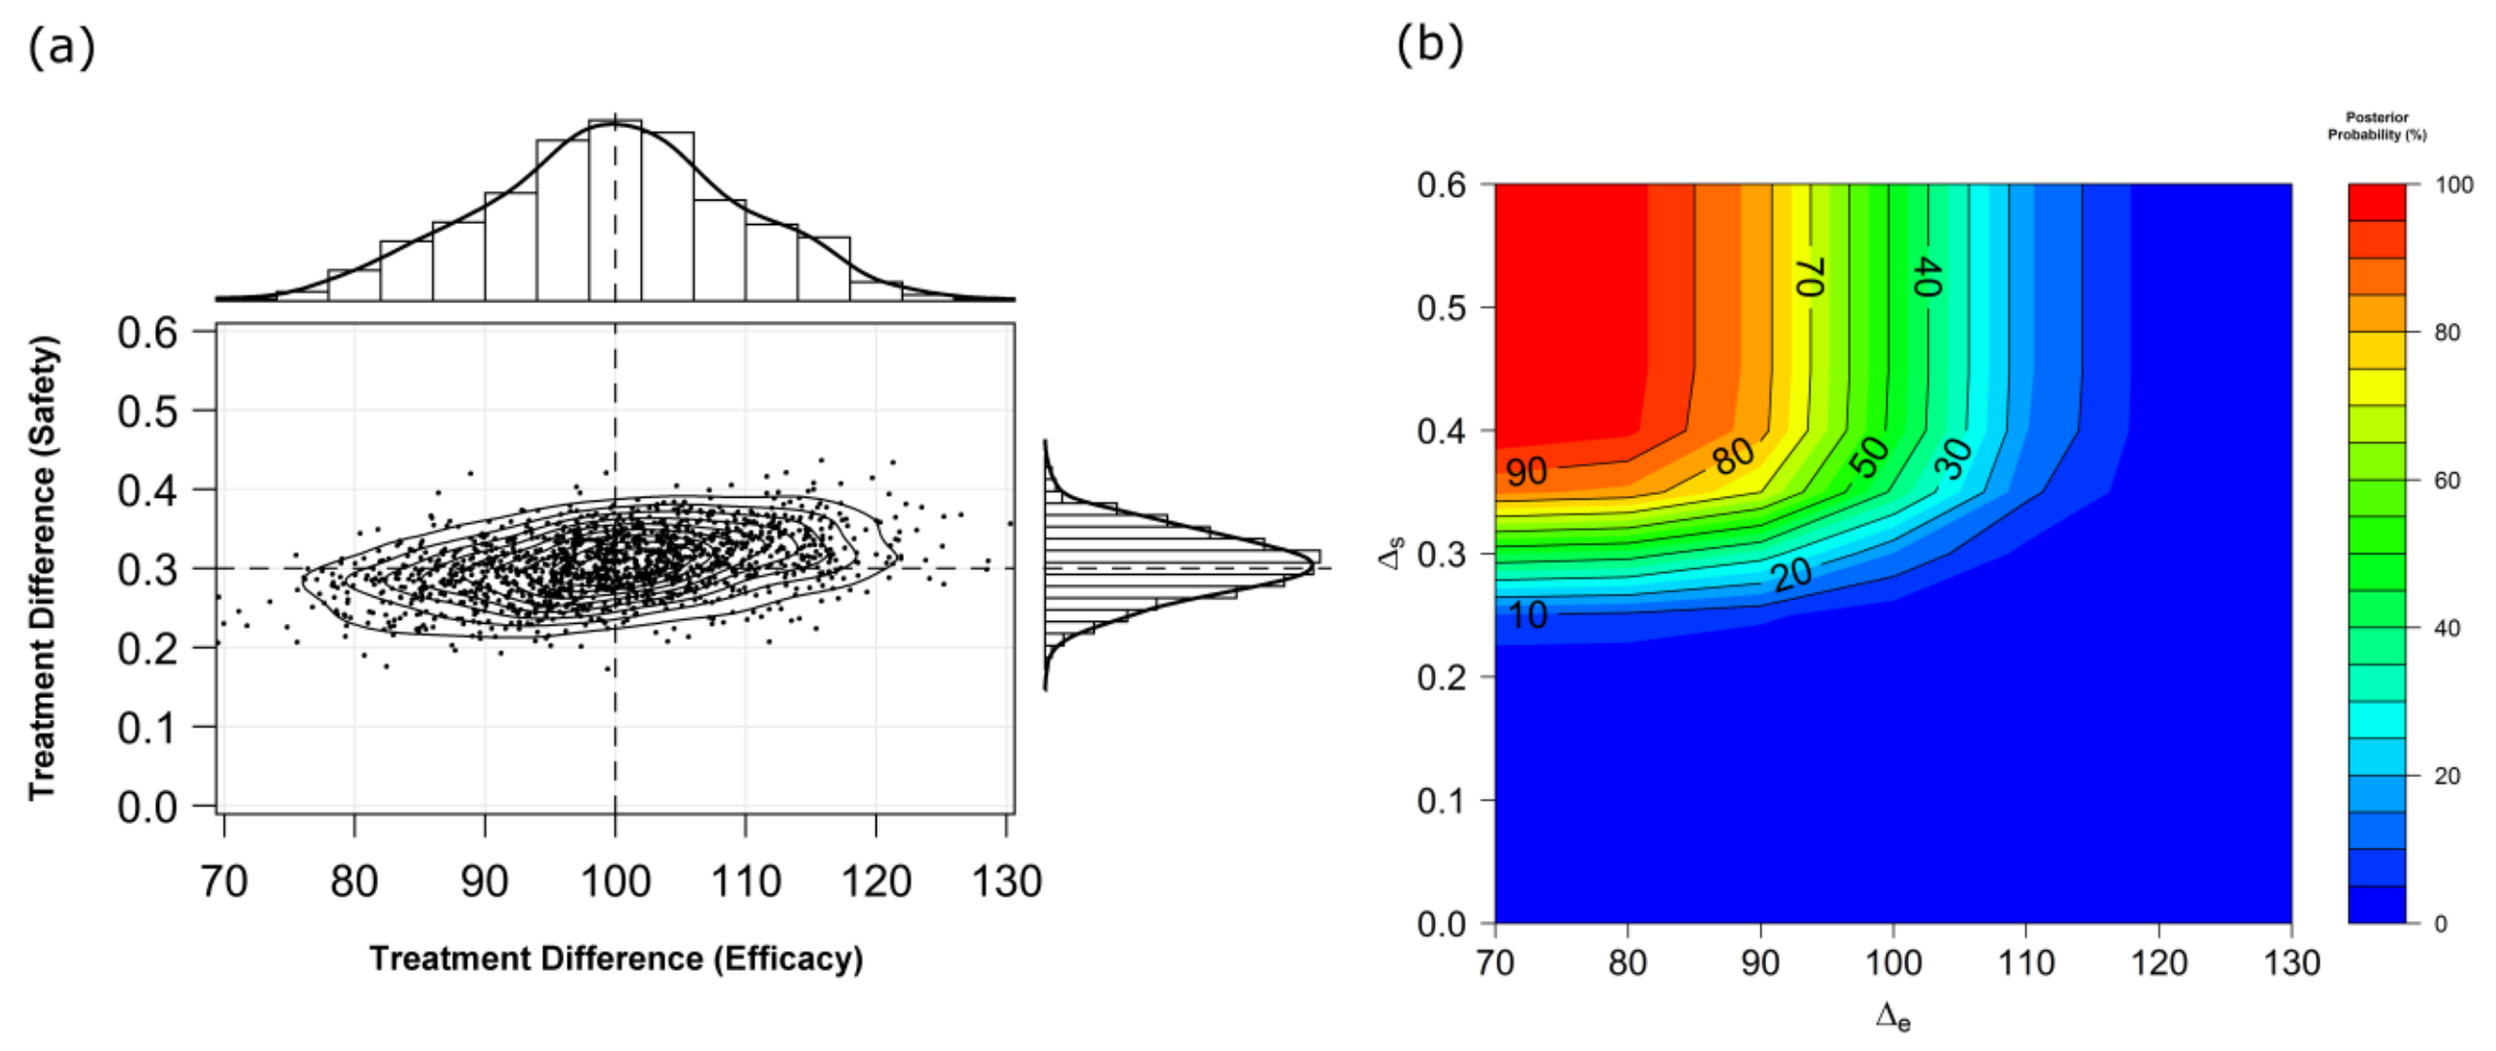
(a)
(b)
Treatment Difference (Safety)
Treatment Difference (Efficacy)
Posterior Probability (%)

Figure 1. Single simulated example
(a) Joint posterior distribution for $H_E(\mu_1,\ \mu_2) = \mu_2 - \mu_1$ and $H_S(p_1,\ p_2) = p_2 - p_1$.
(b) Contour plot showing the POTS: P($H_E(\mu_1,\ \mu_2\) \ \geq\ \Delta_E$ and $H_S(p_2\ ,\ p_1) \ \leq\ \Delta_S$| Data) for values of Δ_E and Δ_S. Each contour has constant posterior probability.

Simulation Study 2: Dose Response

- The copula methodology was applied to the problem of optimal dose selection in Phase 2.
- An efficacy Emax model was combined with a standard non-linear regression for a binary safety event using the Gaussian copula allowing for increased dependency between the responses as the dose increased.
- Using definitions of **minimum effective dose (MED)** and **critical effective dose (CED)** viable dose ranges can be represented graphically to assess possible Phase 3 doses.
- Simulations for clinical trial design investigated the power to achieve specific BR profiles given characteristics such as sample size, efficacy effect, safety effect and dependence between responses.

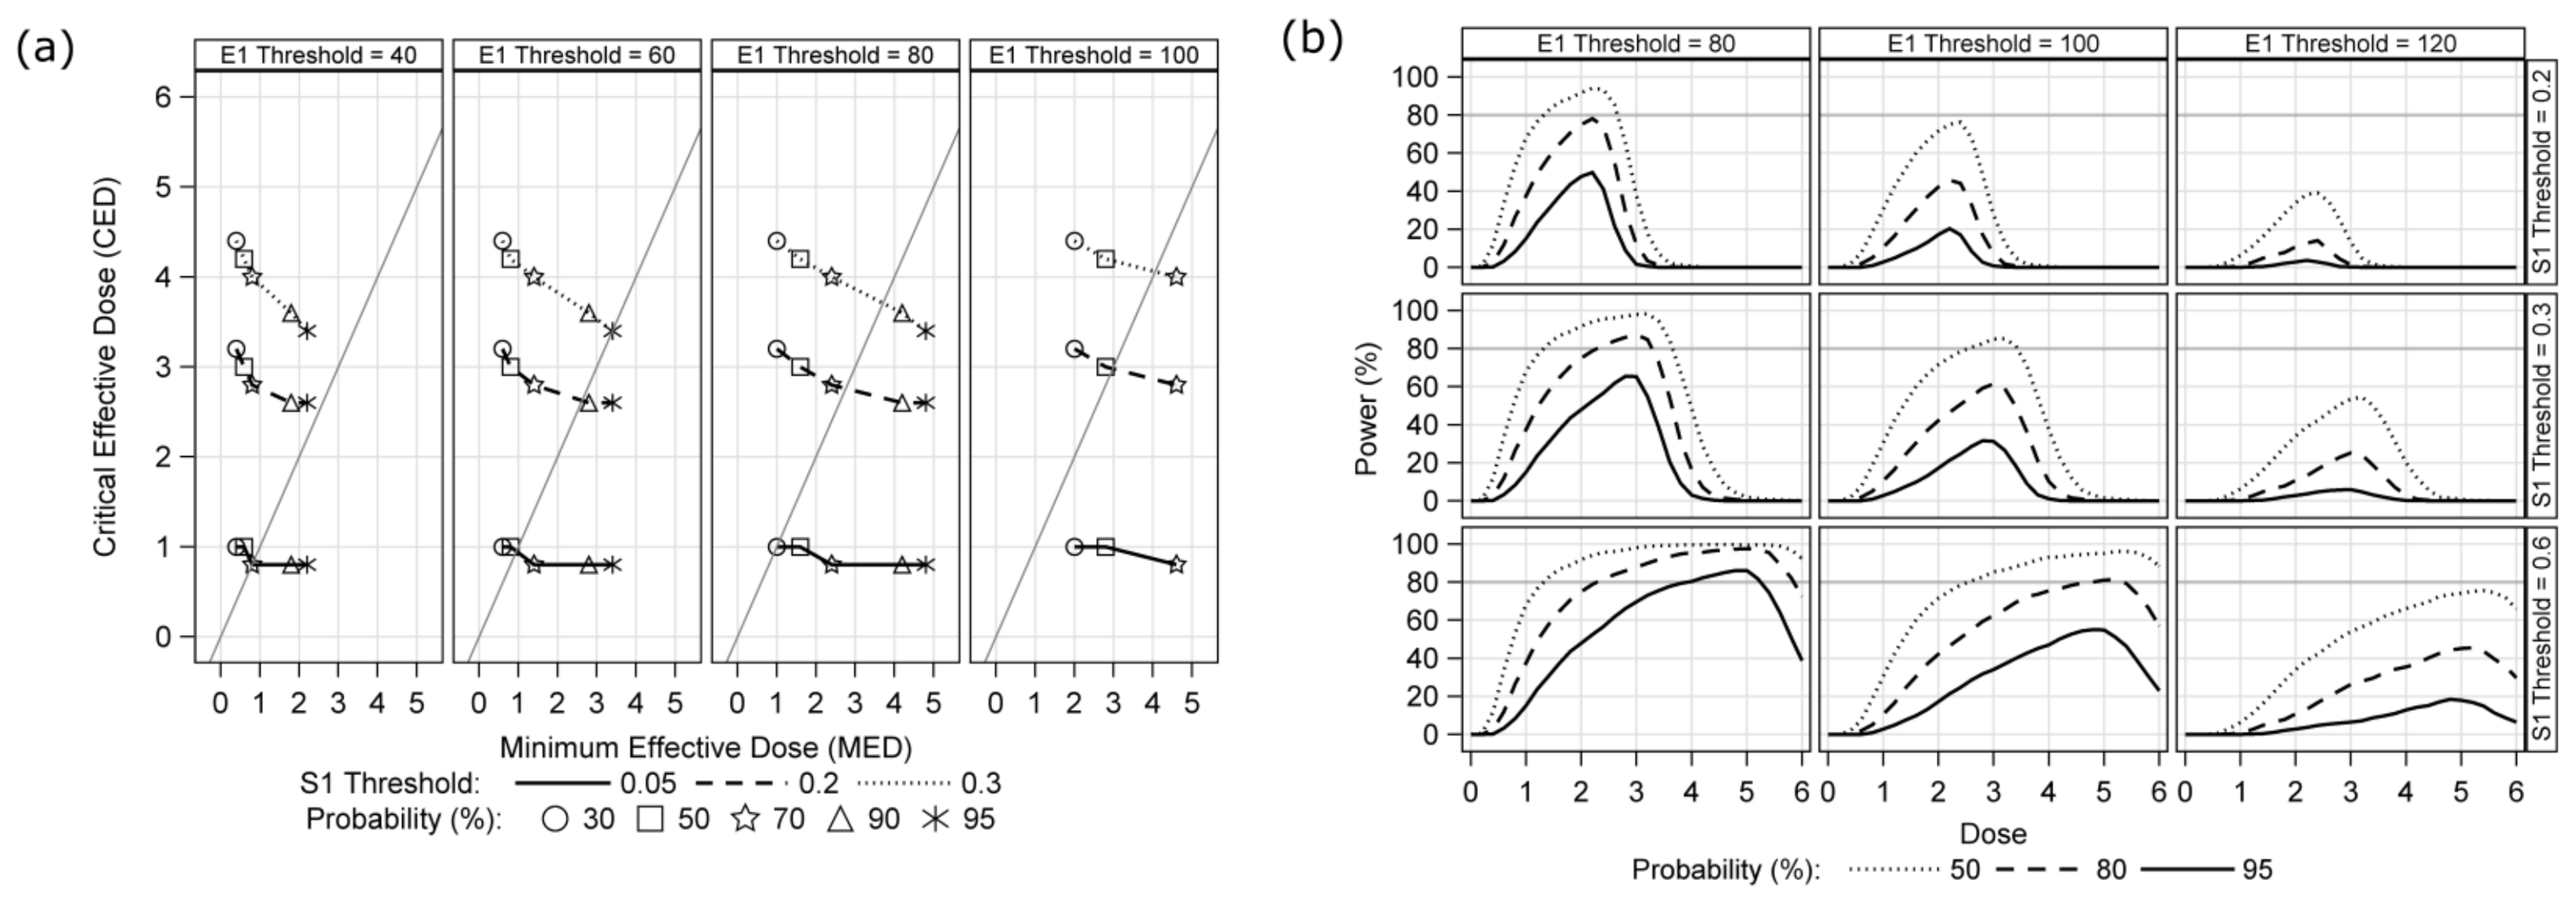
(a)
E1 Threshold = 40
E1 Threshold = 60
E1 Threshold = 80
E1 Threshold = 100
Critical Effective Dose (CED)
Minimum Effective Dose (MED)
S1 Threshold: 0.05 0.2 0.3
Probability (%): 30 50 70 90 95
(b)
E1 Threshold = 80
E1 Threshold = 100
E1 Threshold = 120
S1 Threshold = 0.2
S1 Threshold = 0.3
S1 Threshold = 0.6
Power (%)
Dose
Probability (%): 50 80 95

Figure 2.
(a) MED vs CED for values of Δ_E (Efficacy Threshold), Δ_S (Safety Threshold) and p (Probability, %) for a sample simulated dataset. Diagonal represents the MED=CED line.
(b) Power vs dose for different BR scenarios. Dose-response data for efficacy and safety modelled jointly using Gaussian copula.

Real Data Example: Diabetes

- A clinical trial was conducted to assess the efficacy and safety of a potential new treatment for patients with new-onset type 1 diabetes over an 18-month period.
- The primary efficacy endpoint was **change from baseline in C-peptide levels at 6 months** and adverse events of interest included **infection** and **Cytokine Release Syndrome (CRS)**. In total 73 subjects had C-peptide levels recorded at 6 months (39 received active vs 34 placebo). Further details of the study have been anonymised.
- An initial BR analysis was conducted on the primary efficacy and infection endpoints using GLMMs and included the impact of baseline C-peptide levels.
- Further analysis of the primary efficacy endpoint, infections and CRS were carried out using CMR. Joint posterior POTS values were presented for various thresholds of the three responses.

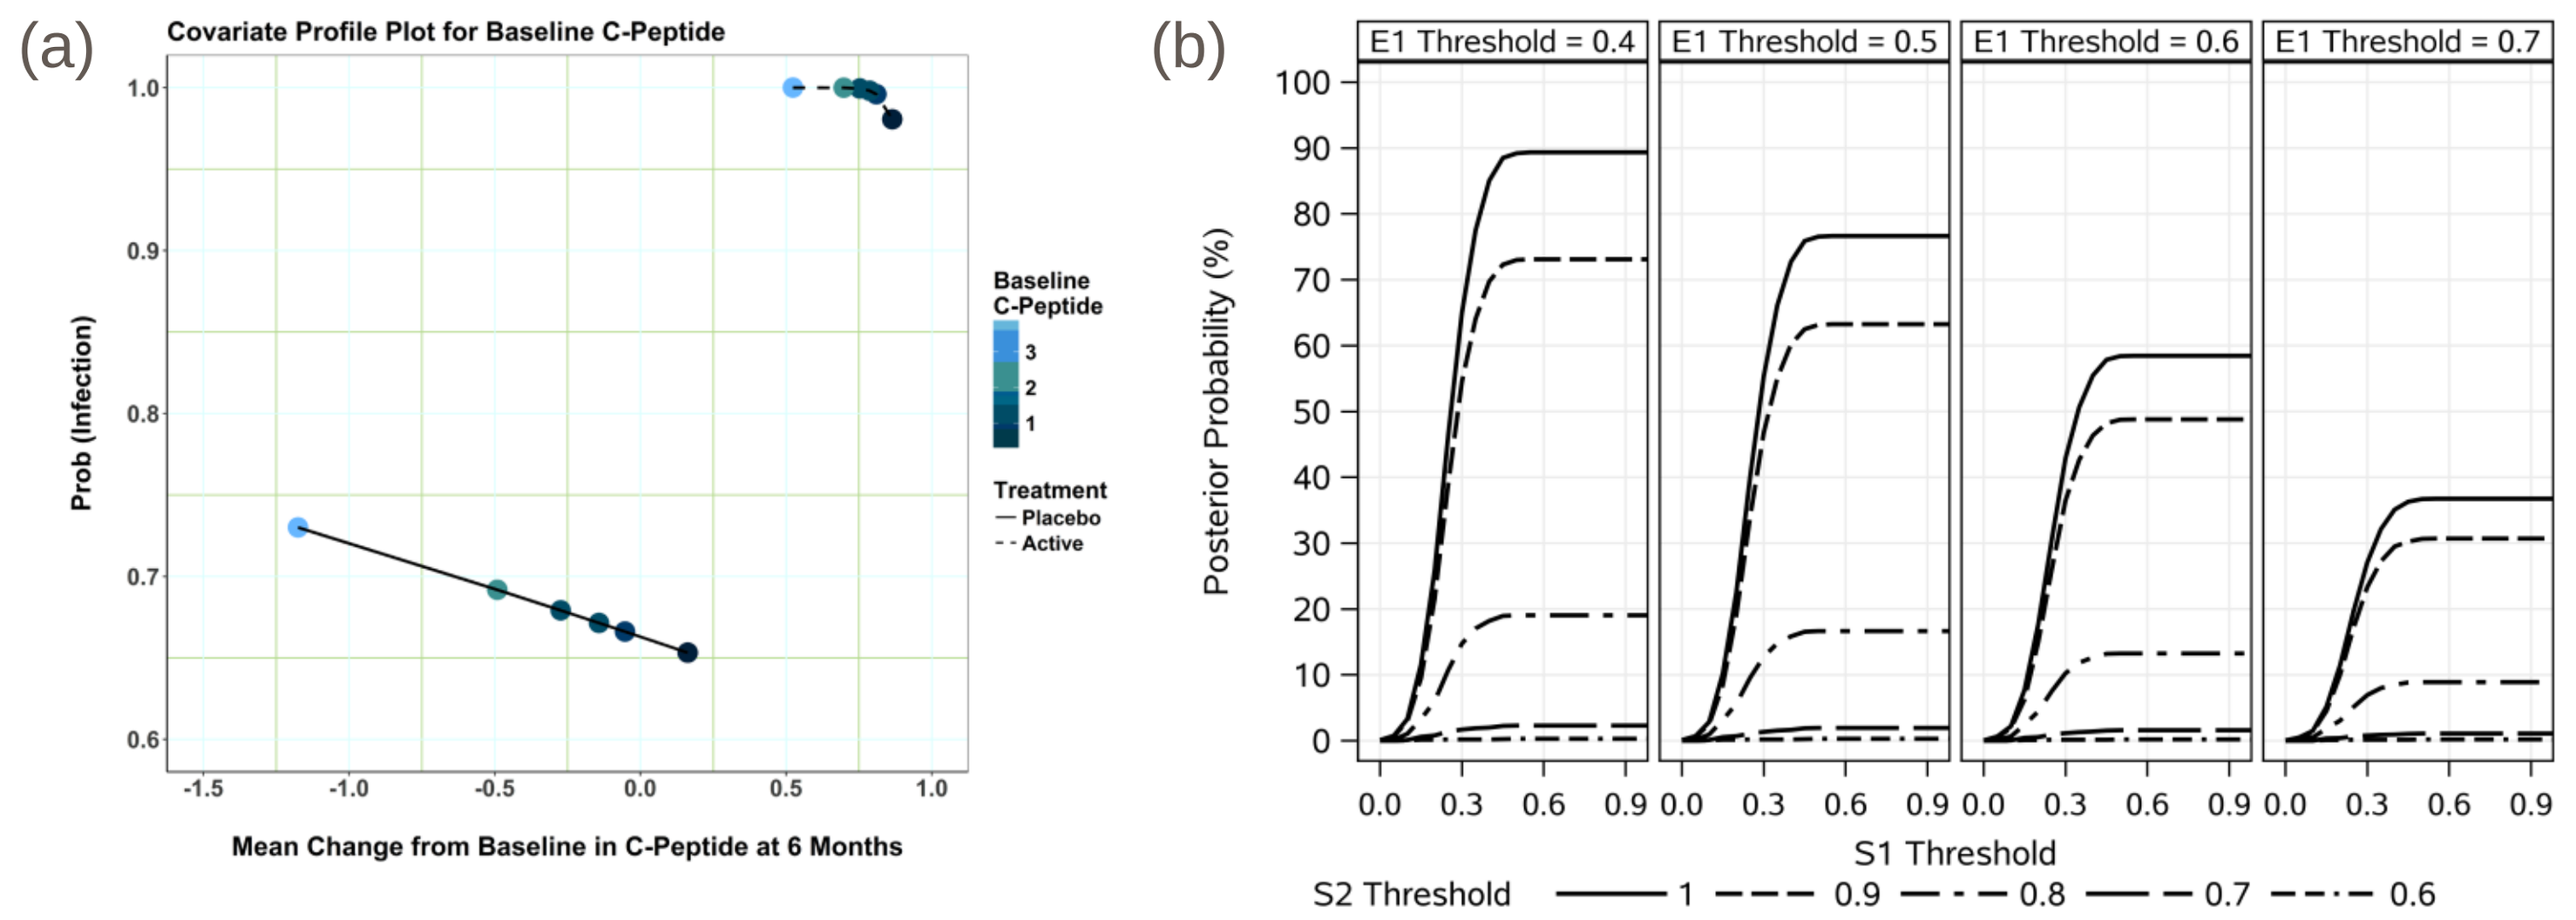
(a)
Covariate Profile Plot for Baseline C-Peptide
Prob (Infection)
Mean Change from Baseline in C-Peptide at 6 Months
Baseline C-Peptide
Treatment
Placebo
Active
(b)
E1 Threshold = 0.4
E1 Threshold = 0.5
E1 Threshold = 0.6
E1 Threshold = 0.7
Posterior Probability (%)
S1 Threshold
S2 Threshold 1 0.9 0.8 0.7 0.6

Figure 3:
(a) Covariate profile plot with predicted change in BR for active and placebo given different baseline C-Peptide levels.
(b) Posterior POTS for primary efficacy, infections and CRS risks (active vs. placebo) for various values of Δ_{E1}, Δ_{S1} and Δ_{S2}.

Conclusions

- **Joint modelling allowed more accurate estimates of the BR profile, especially when the responses exhibited large dependency.**
- **Communicating BR results to different stakeholders using Bayesian decision making criteria and clinically meaningful thresholds is simple and intuitive.**
- **Both subject-specific and population average estimates can be assessed, enabling different stakeholders to focus on the estimate that is most relevant for their purpose.**
- **Beyond 3 dimensions modelling dependence and visualizing the outcome can be challenging so alternative BR methodologies could be considered.**
- **Joint modelling of BR can be applied at the clinical trial design stage but this may be resource intensive.**

Acknowledgements

The authors would like to thank, Graeme Archer, Nicky Best, Nigel Dallow and James Roger for useful discussions and GSK for initially sponsoring the work which is now published.

Costa MJ, Drury T. Bayesian joint modelling of benefit and risk in drug development. Pharmaceutical Statistics. 2018;17:248–263.

Created by: **Digital Media Services**